Breast cancer-specific survival in patients with 21-gene Recurrence Score® results <11 classified as prognostic stage IA by new 8th edition AJCC Staging Manual

Frederick L. Baehner,[1,2] Valentina I. Petkov,[3] Debbie McCullough,[1] Steven Shak[1]

[1]Genomic Health, Inc., Redwood City, CA; [2]University of California, San Francisco, San Francisco, CA; [3]National Cancer Institute, Bethesda, MD

Background

- Since 1977, the American Joint Committee on Cancer (AJCC) Cancer Staging Manual has been the most comprehensive guide to cancer staging.
- The AJCC Cancer Staging Manual 8th edition (2016) added for the first time histologic grade, ER, PR, and HER2 status, and the 21-gene Oncotype DX Breast Recurrence Score® (RS) assay results for prognostic staging [1].
 - Patients with node-negative (N0), ER+, HER2-negative breast cancer and RS <11 are in prognostic stage IA, regardless of tumor size (T1-T2) or histologic grade (1-3) [1].
 - The RS <11 cutpoint was selected based on results of the TAILORx trial, which showed that patients with RS <11 had a 5-year rate of freedom from distant recurrence of 99.3% [2].
- The SEER Program of the National Cancer Institute is an authoritative source of cancer incidence and survival statistics, collecting population-based data for ~30% of the US [3].
- In a previous analysis of SEER patients with N0 disease, breast cancer-specific survival (BCSS) was assessed by both standard (RS <18, 18-30, ≥31) and TAILORx (RS <11, 12-25, >25) RS cutpoints [4,5]. Patients with N0 disease and RS <11 or 11-17 had favorable 5-year BCSS, even with limited chemotherapy (CT) use.
- Here, we characterize the BCSS of patients with N0, HR+, HER2-negative breast cancer in the SEER registry who have RS <11 across categories of clinicopathologic features.

Objective

- To determine BCSS of SEER patients with hormone receptor-positive (HR+), HER2-negative, N0 invasive breast cancer and RS <11, by categories of age, tumor size, and tumor grade

Methods

- RS results were provided electronically to SEER by Genomic Health as mandated by registry operations [2].
- Patients with RS results were eligible if they had N0, HR+ (by SEER and RT-PCR), HER2-negative (by RT-PCR only; HER2 status not available in SEER prior to 2010) breast cancer, diagnosed between January 2004 and December 2012.
 - Excluded were those with node-positive disease, prior invasive tumors, or concurrent multiple tumors.
- SEER demographics, tumor characteristics, reported CT use, and BCSS were available through 2013.
 - Under-reporting of CT use to SEER is well-known [6].
- BCSS was defined according to pre-existing robust methodology [7]. Actuarial estimates of BCSS by reported CT use were computed through 5 years with 95% confidence intervals (CI) using standard and nonstandard cutpoints (RS <11, 11-17, 18-25, 26-30, ≥31). The log-rank test was used to compare across RS groups.

Results

Figure 1. Total SEER Population and Patients With N0, HR+, HER-negative Disease Eligible for This Analysis

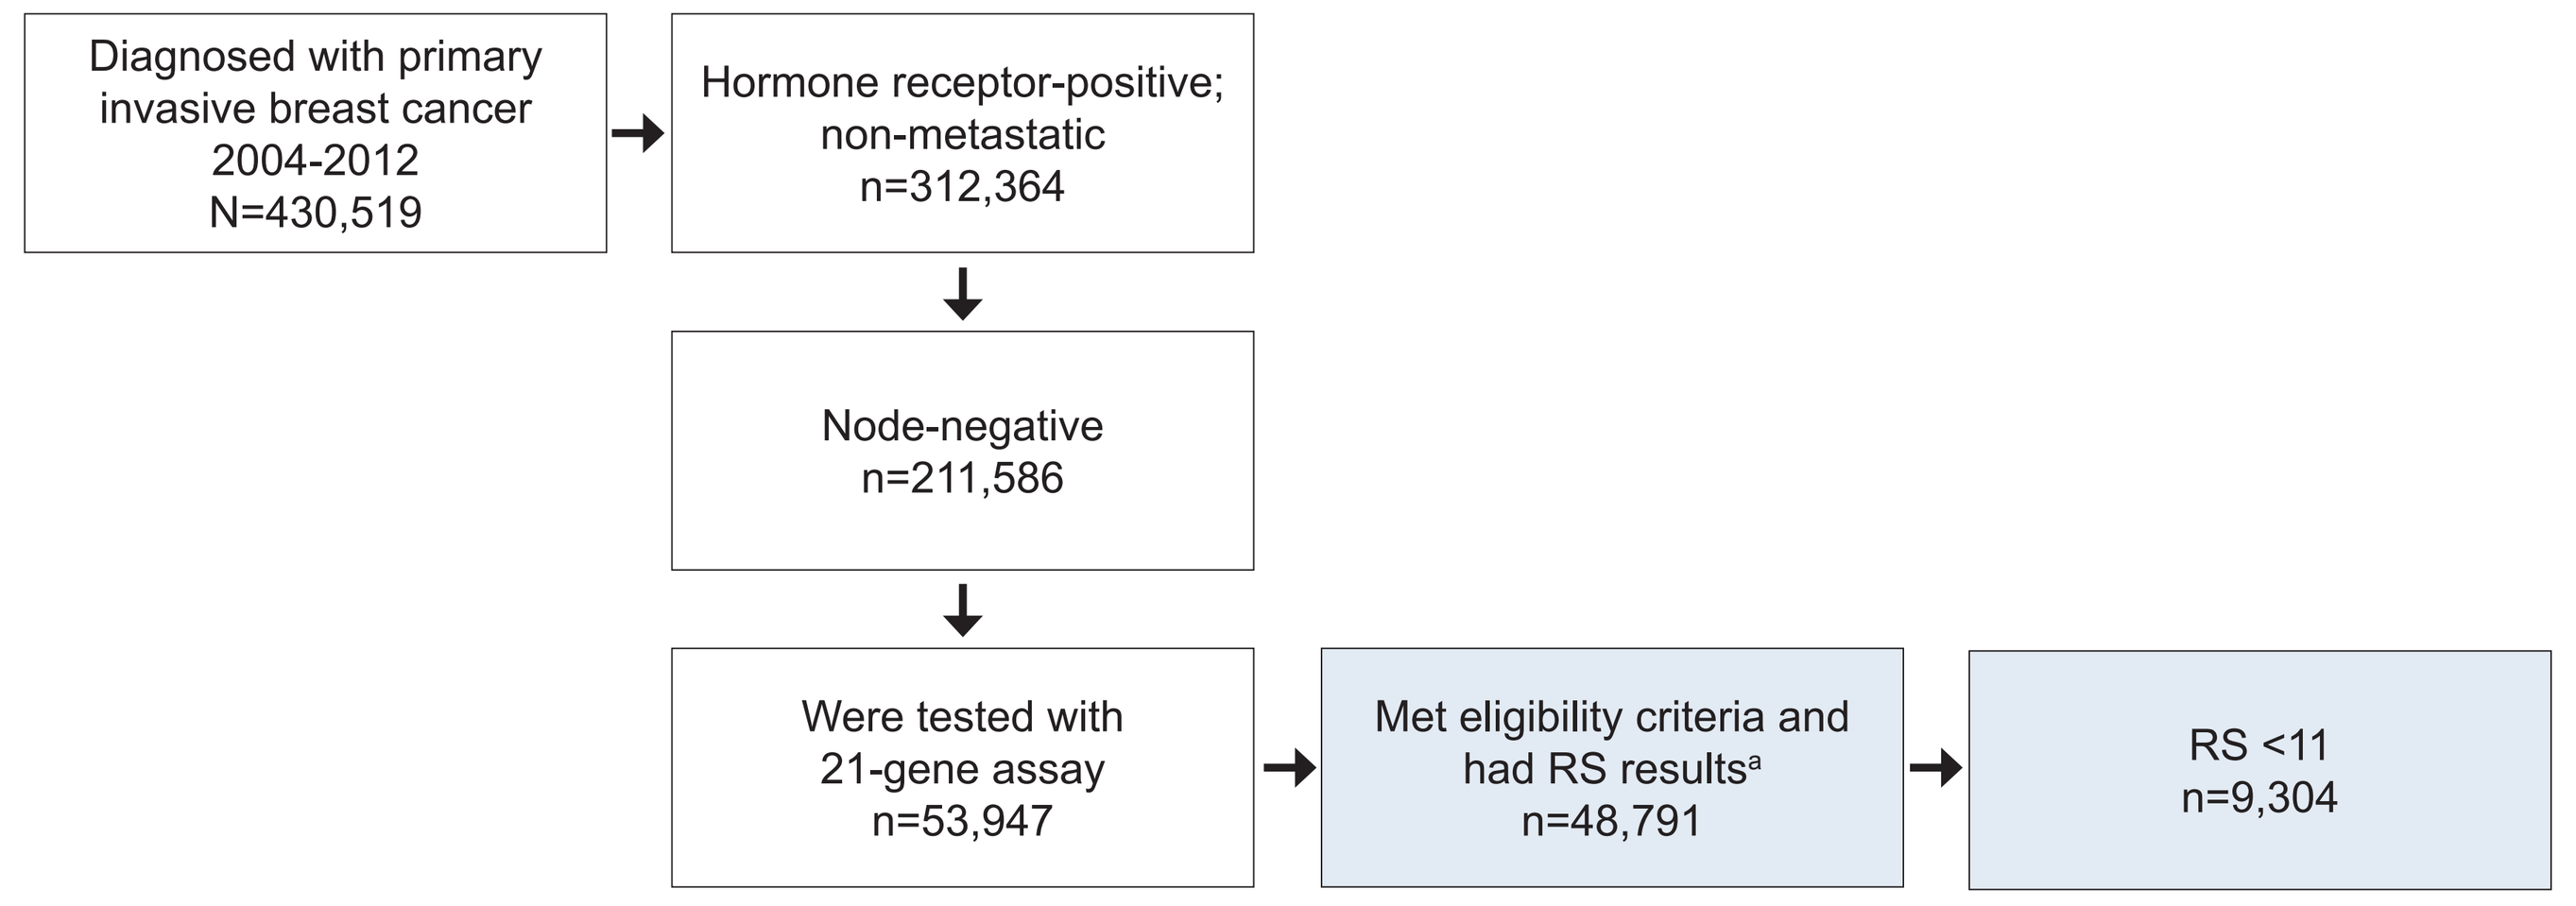
Diagnosed with primary invasive breast cancer 2004-2012 N=430,519
Hormone receptor-positive; non-metastatic n=312,364
Node-negative n=211,586
Were tested with 21-gene assay n=53,947
Met eligibility criteria and had RS results[a] n=48,791
RS <11 n=9,304

a. The cohort that met eligibility requirements, was tested, and had RS results excludes 4,266 patients (8% of 53,947 patients) for the following reasons: HER2-positive breast cancer based on 21-gene assay HER2 single-gene result (3%), inadequate tissue sample (2%), multiple tumors (2%), and test cancellation (1%). Median follow-up overall was 44 months, median follow-up for RS <11 was 40 months.

Results

Table 1. Patient and Tumor Characteristics

		RS <11 (N=9304)
Age at diagnosis, y	≤50	2299 (24.7%)
	51 to 70	5756 (61.9%)
	>70	1249 (13.4%)
Tumor size[a], cm	≤1	1808 (19.5%)
	>1 to 2	5056 (54.7%)
	>2 to 3	1734 (18.8%)
	>3 to 4	445 (4.8%)
	≥4	203 (2.2%)
Grade[a,b]	Low	3426 (38.0%)
	Intermediate	4909 (54.4%)
	High	686 (7.6%)
Race[a]	White	7747 (83.8%)
	Black	704 (7.6%)
	Other	797 (8.6%)
SES quintile[a] (Yost)	Lowest	1144 (12.5%)
	2nd lowest	1439 (15.7%)
	Middle	1757 (19.2%)
	2nd highest	2084 (22.7%)
	Highest	2749 (30.0%)

a. Among patients with nonmissing information. b. Low=well differentiated; intermediate=moderately differentiated; high=poorly differentiated, undifferentiated, or anaplastic.

- A total of 9,304 patients with N0, HR+, HER2-negative breast cancer in the SEER also had RS <11 (19% of those who met all other eligibility criteria).
- There was a wide range of ages, tumor size, and tumor grade.
- Median age was 59 years.
- Tumor size was >2 cm in 21% of patients.
- Histologic tumor grade was intermediate (moderately differentiated) and high (poorly differentiated) in 54% and 8% of patients, respectively.
- Median follow-up for this cohort was 40 months.

Figure 2. Distribution of Patients With RS <11 by Age, Grade, and Tumor Size

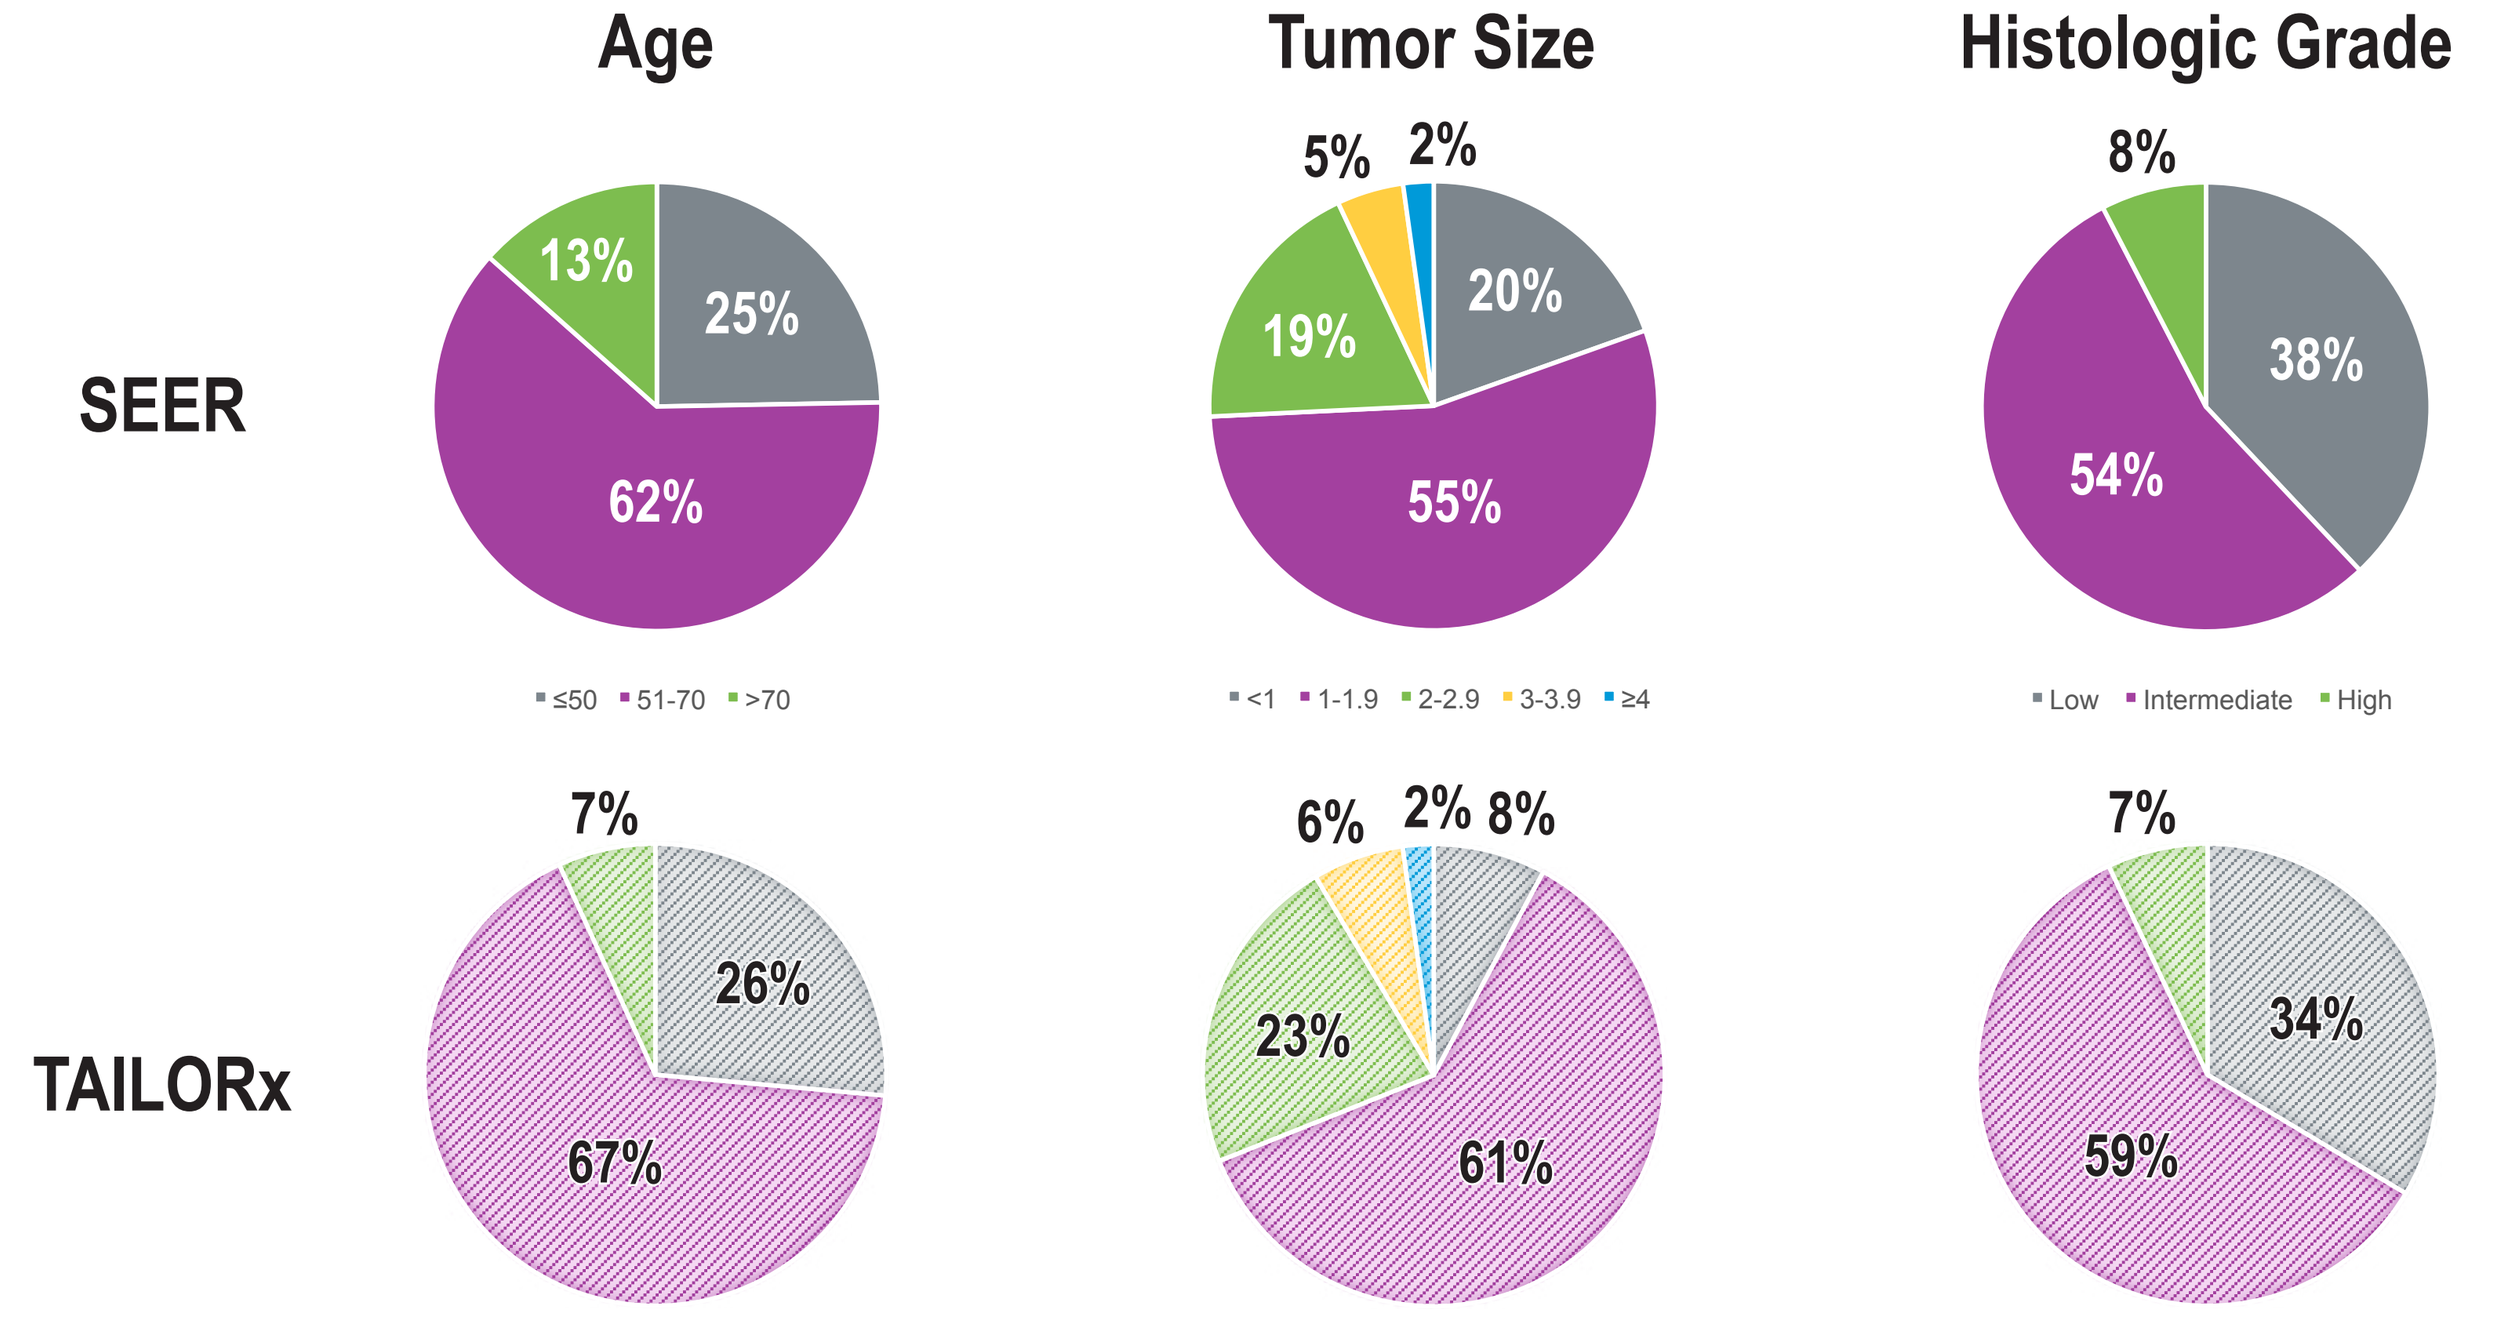
Age
Tumor Size
Histologic Grade
SEER
TAILORx
13%
25%
62%
≤50 51-70 >70
5% 2%
19%
20%
55%
<1 1-1.9 2-2.9 3-3.9 ≥4
8%
38%
54%
Low Intermediate High
7%
26%
67%
6% 2% 8%
23%
61%
7%
34%
59%

For more information about SEER, visit http://seer.cancer.gov/.
For more information about the Surveillance Research Program, visit http://surveillance.cancer.gov/
@NCICancerStats

Results

Figure 3.
5-year BCSS Overall and RS <11, All Patients

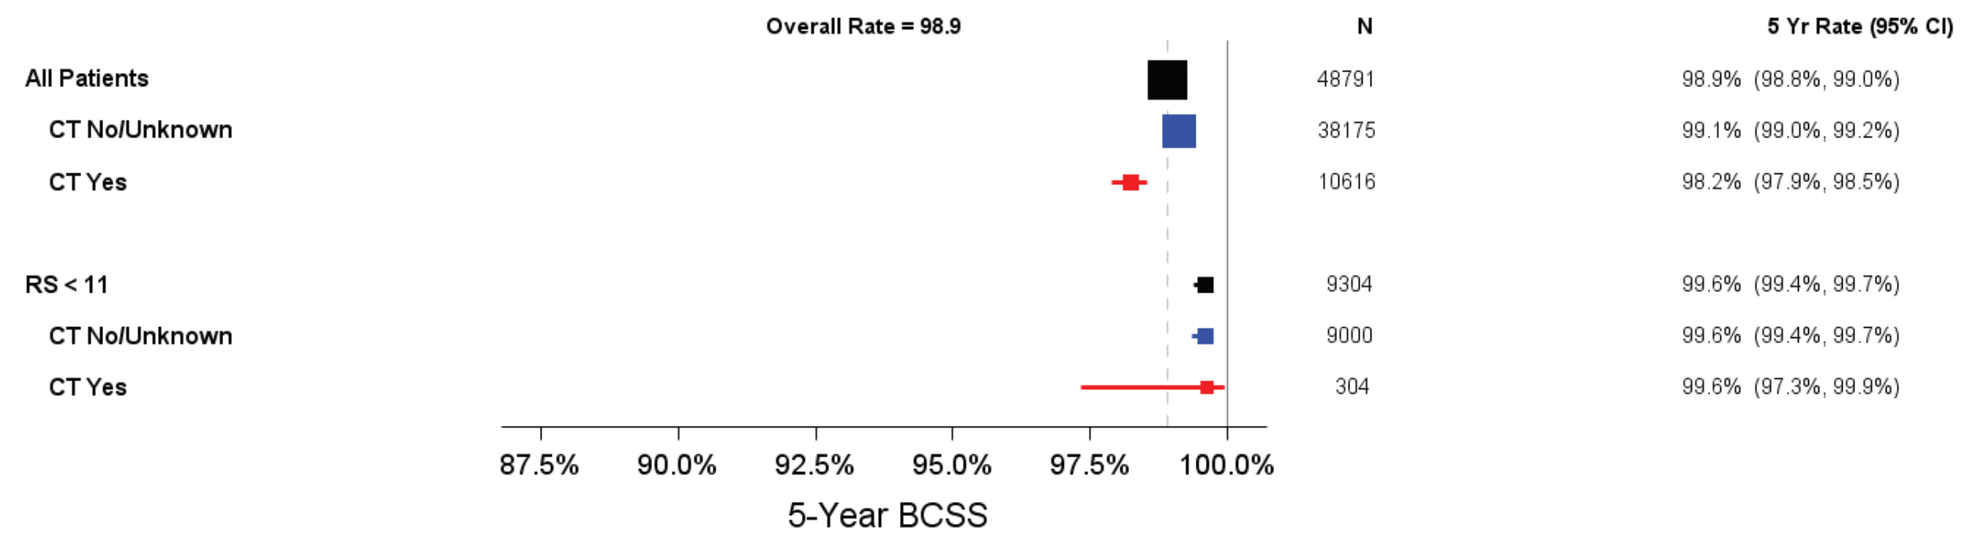
Overall Rate = 98.9
N
5 Yr Rate (95% CI)
All Patients 48791 98.9% (98.8%, 99.0%)
CT No/Unknown 38175 99.1% (99.0%, 99.2%)
CT Yes 10616 98.2% (97.9%, 98.5%)
RS < 11 9304 99.6% (99.4%, 99.7%)
CT No/Unknown 9000 99.6% (99.4%, 99.7%)
CT Yes 304 99.6% (97.3%, 99.9%)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

*CT use is known to be under-reported in the SEER registries.

- 9000 patients with RS <11 and CT reported no/unknown had >99% 5-year BCSS

5-year BCSS Overall and RS <11, by Age

Age ≤50 y

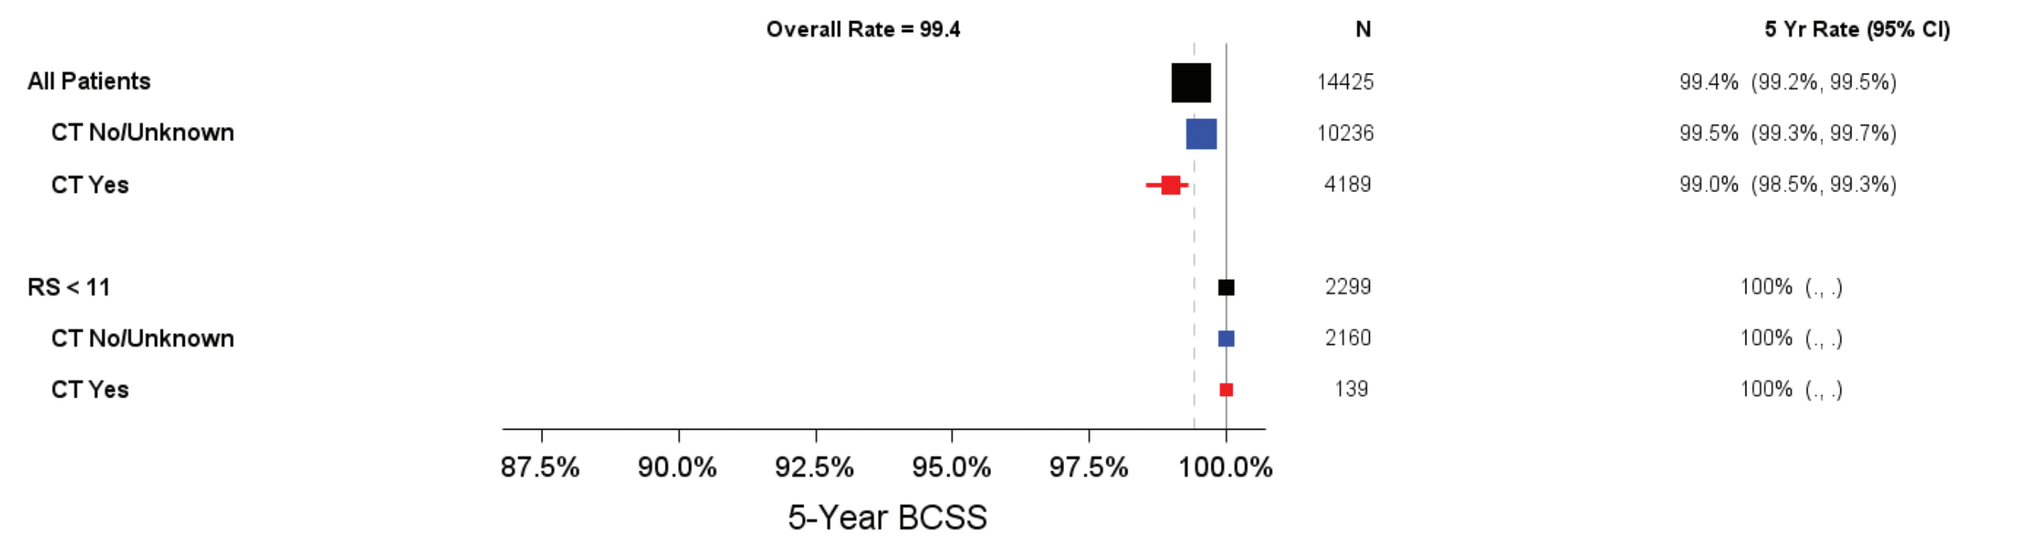
Overall Rate = 99.4
N
5 Yr Rate (95% CI)
All Patients 14425 99.4% (99.2%, 99.5%)
CT No/Unknown 10236 99.5% (99.3%, 99.7%)
CT Yes 4189 99.0% (98.5%, 99.3%)
RS < 11 2299 100% (., .)
CT No/Unknown 2160 100% (., .)
CT Yes 139 100% (., .)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Age 51-70 y

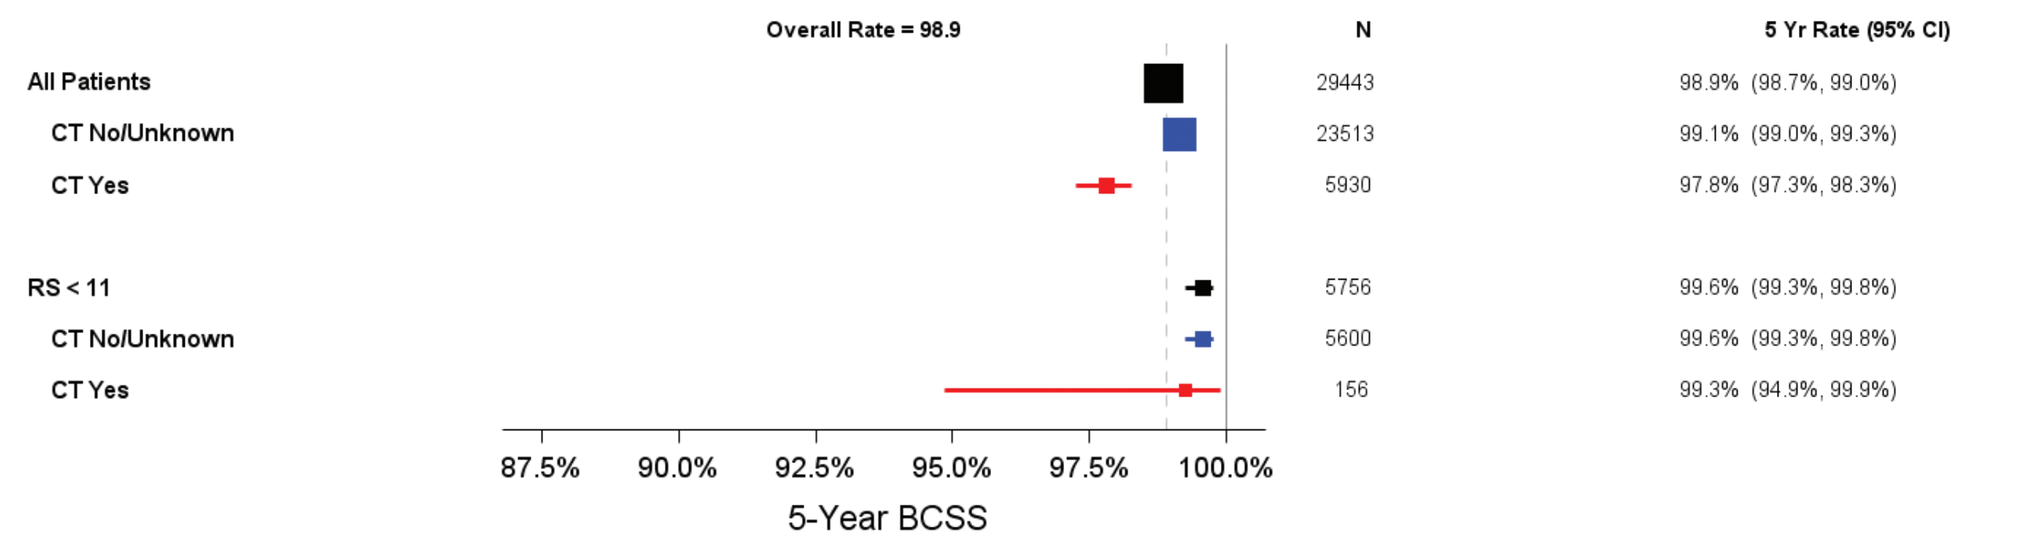
Overall Rate = 98.9
N
5 Yr Rate (95% CI)
All Patients 29443 98.9% (98.7%, 99.0%)
CT No/Unknown 23513 99.1% (99.0%, 99.3%)
CT Yes 5930 97.8% (97.3%, 98.3%)
RS < 11 5756 99.6% (99.3%, 99.8%)
CT No/Unknown 5600 99.6% (99.3%, 99.8%)
CT Yes 156 99.3% (94.9%, 99.9%)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Age >70 y

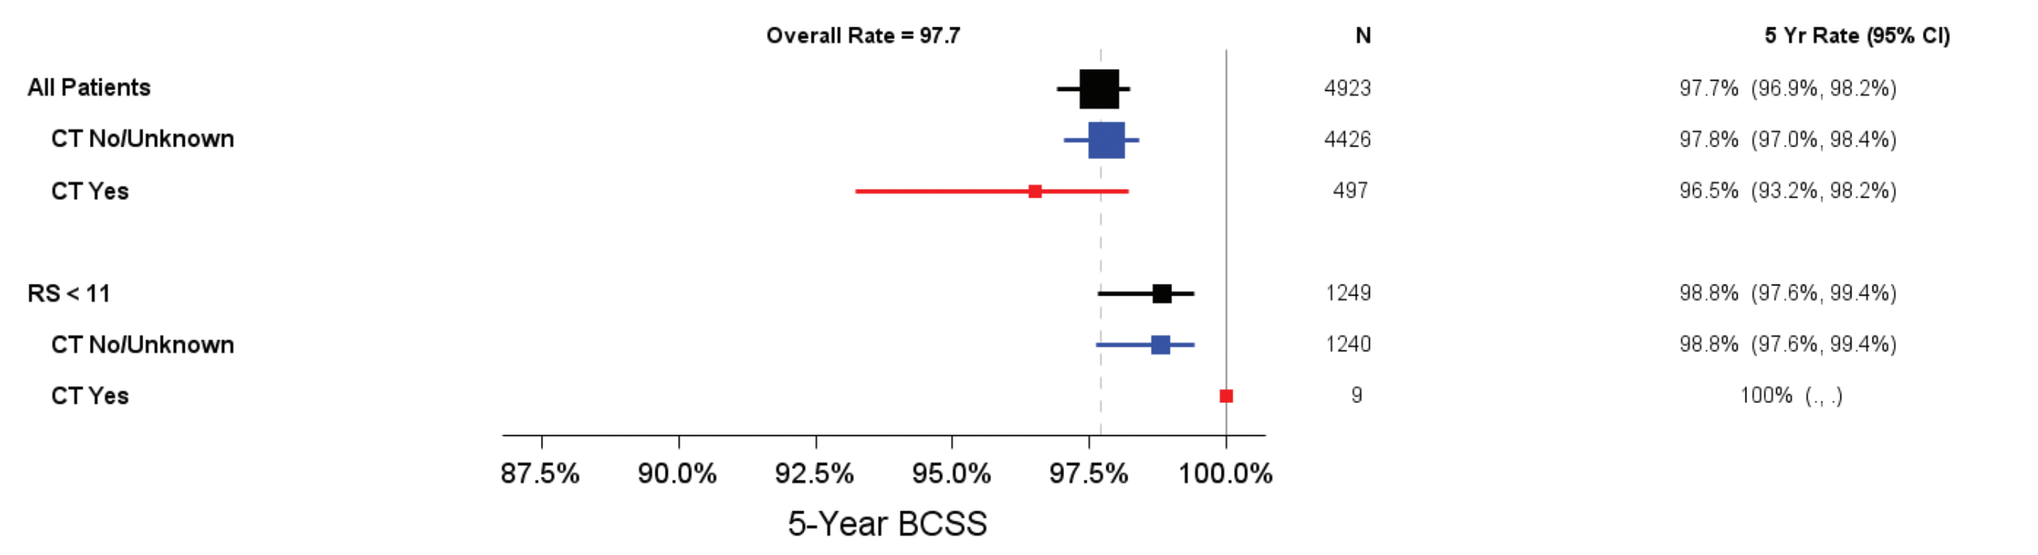
Overall Rate = 97.7
N
5 Yr Rate (95% CI)
All Patients 4923 97.7% (96.9%, 98.2%)
CT No/Unknown 4426 97.8% (97.0%, 98.4%)
CT Yes 497 96.5% (93.2%, 98.2%)
RS < 11 1249 98.8% (97.6%, 99.4%)
CT No/Unknown 1240 98.8% (97.6%, 99.4%)
CT Yes 9 100% (., .)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

5-year BCSS Overall and RS <11, by Grade

Low Grade

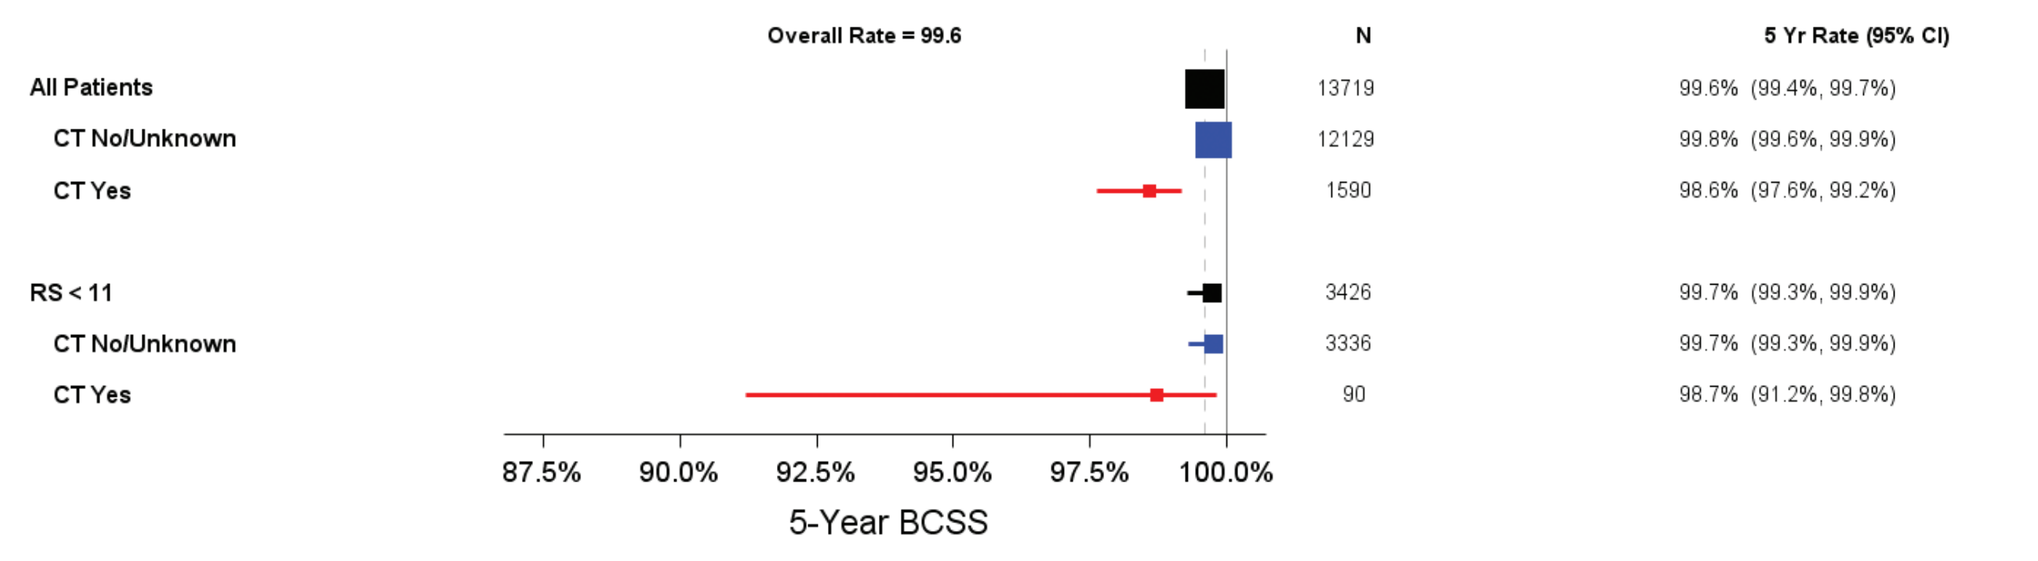
Overall Rate = 99.6
N
5 Yr Rate (95% CI)
All Patients 13719 99.6% (99.4%, 99.7%)
CT No/Unknown 12129 99.8% (99.6%, 99.9%)
CT Yes 1590 98.6% (97.6%, 99.2%)
RS < 11 3426 99.7% (99.3%, 99.9%)
CT No/Unknown 3336 99.7% (99.3%, 99.9%)
CT Yes 90 98.7% (91.2%, 99.8%)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Intermediate Grade

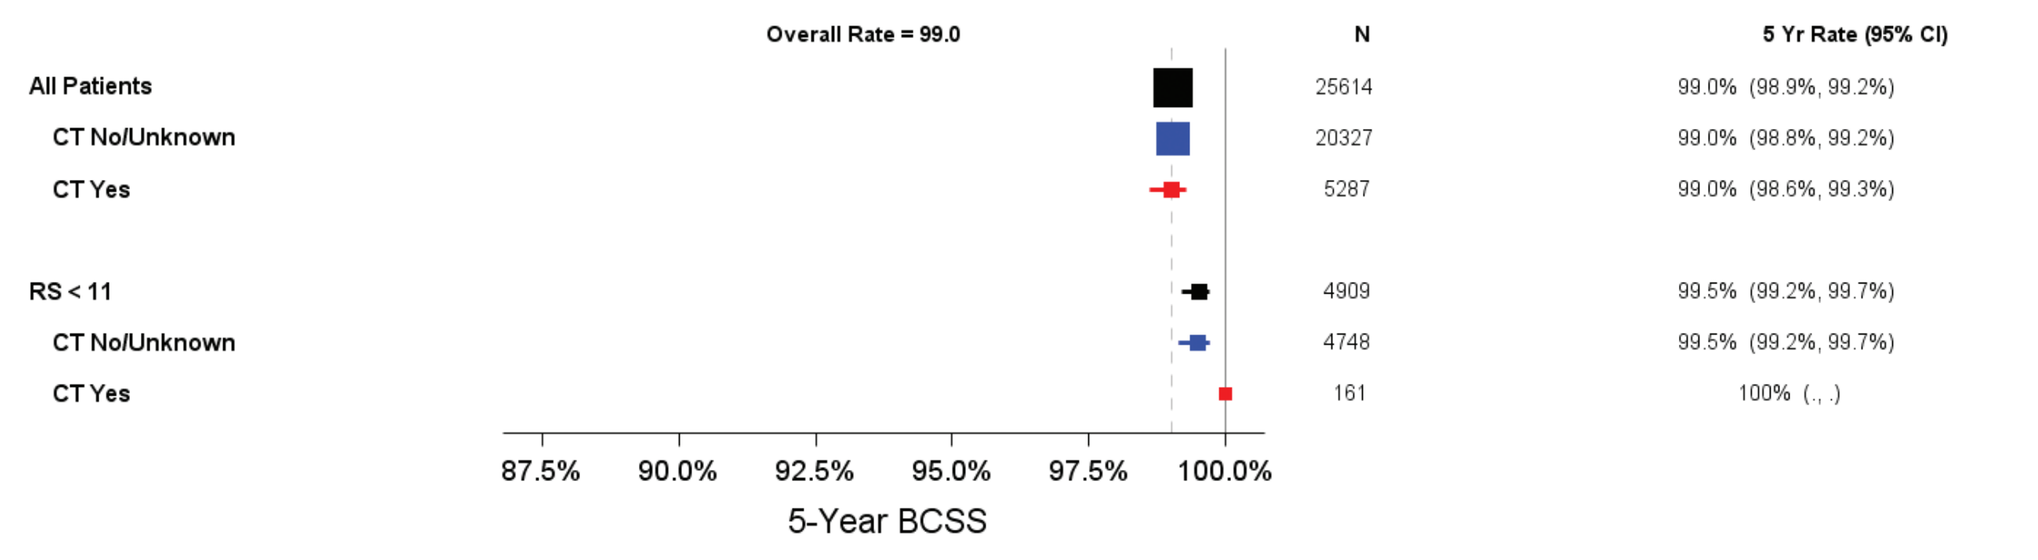
Overall Rate = 99.0
N
5 Yr Rate (95% CI)
All Patients 25614 99.0% (98.9%, 99.2%)
CT No/Unknown 20327 99.0% (98.8%, 99.2%)
CT Yes 5287 99.0% (98.6%, 99.3%)
RS < 11 4909 99.5% (99.2%, 99.7%)
CT No/Unknown 4748 99.5% (99.2%, 99.7%)
CT Yes 161 100% (., .)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

High Grade

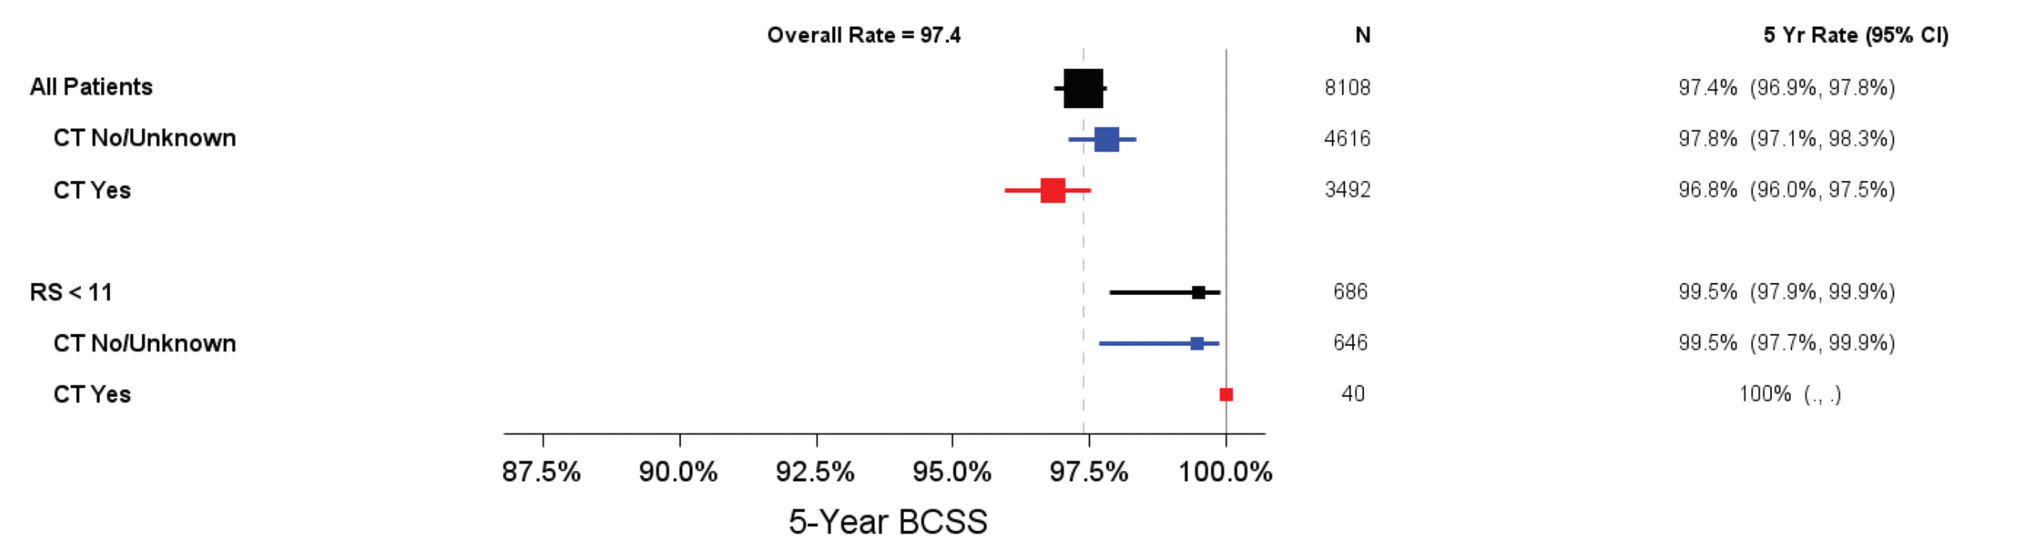
Overall Rate = 97.4
N
5 Yr Rate (95% CI)
All Patients 8108 97.4% (96.9%, 97.8%)
CT No/Unknown 4616 97.8% (97.1%, 98.3%)
CT Yes 3492 96.8% (96.0%, 97.5%)
RS < 11 686 99.5% (97.9%, 99.9%)
CT No/Unknown 646 99.5% (97.7%, 99.9%)
CT Yes 40 100% (., .)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Results

5-year BCSS Overall and RS <11, by Tumor Size

Size ≤1 cm

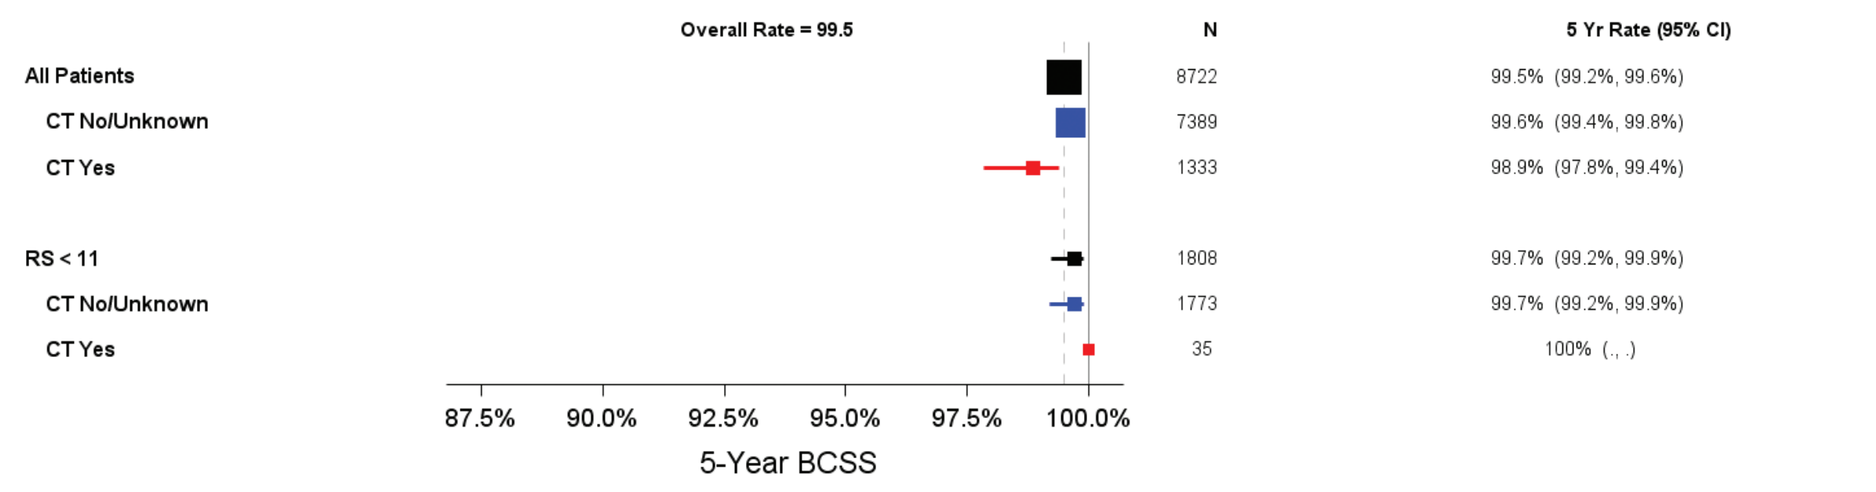
Overall Rate = 99.5
N
5 Yr Rate (95% CI)
All Patients 8722 99.5% (99.2%, 99.6%)
CT No/Unknown 7389 99.6% (99.4%, 99.8%)
CT Yes 1333 98.9% (97.8%, 99.4%)
RS < 11 1808 99.7% (99.2%, 99.9%)
CT No/Unknown 1773 99.7% (99.2%, 99.9%)
CT Yes 35 100% (., .)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Size 1.1-2 cm

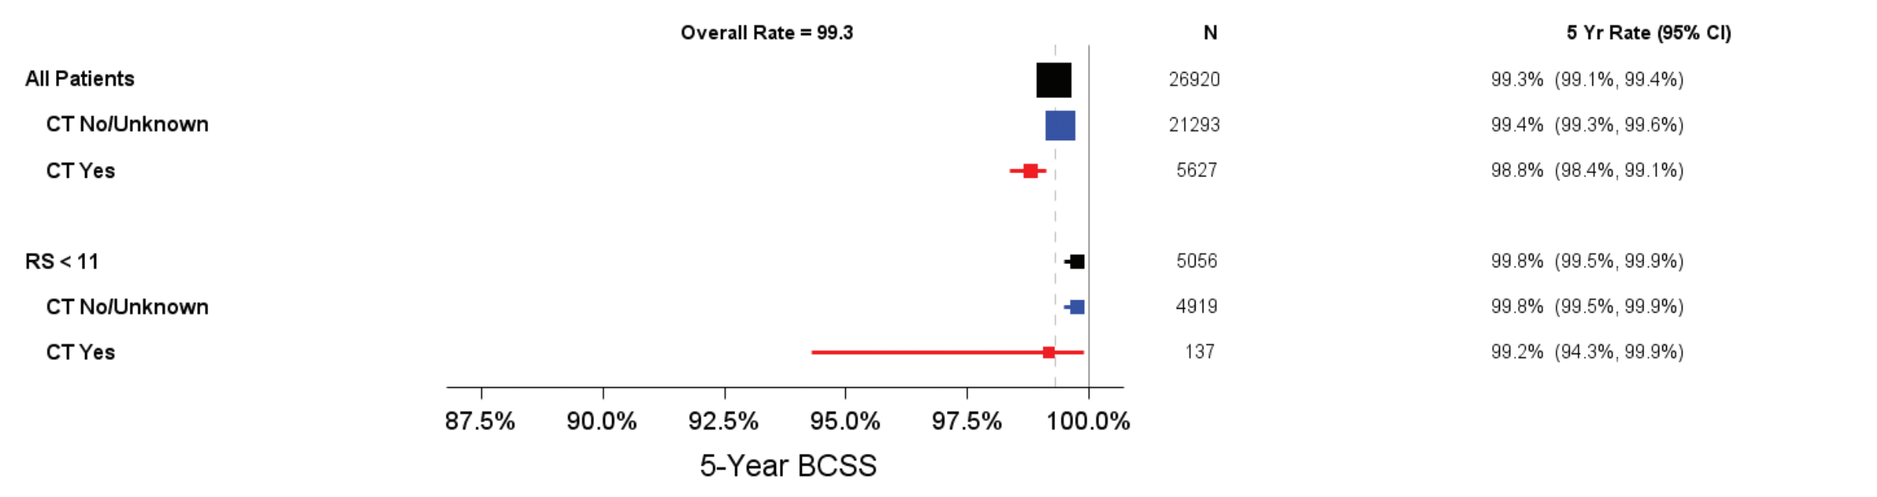
Overall Rate = 99.3
N
5 Yr Rate (95% CI)
All Patients 26920 99.3% (99.1%, 99.4%)
CT No/Unknown 21293 99.4% (99.3%, 99.6%)
CT Yes 5627 98.8% (98.4%, 99.1%)
RS < 11 5056 99.8% (99.5%, 99.9%)
CT No/Unknown 4919 99.8% (99.5%, 99.9%)
CT Yes 137 99.2% (94.3%, 99.9%)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Size 2.1-3 cm

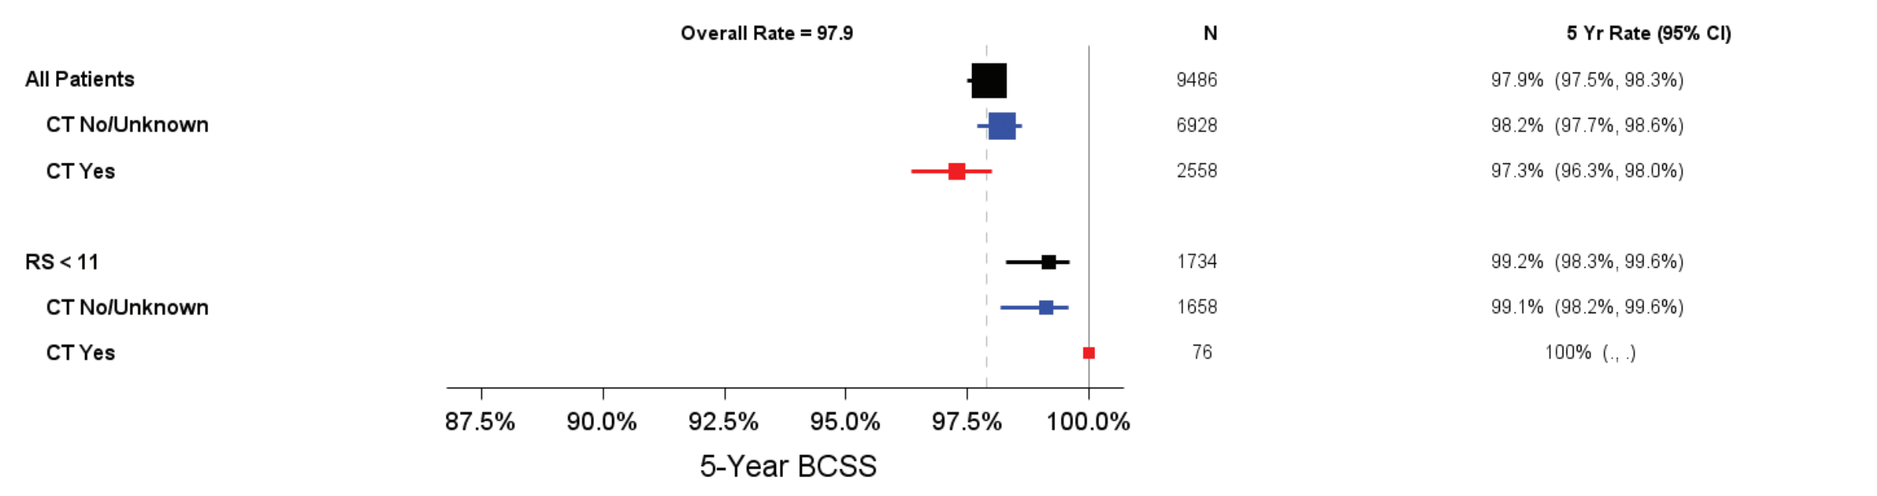
Overall Rate = 97.9
N
5 Yr Rate (95% CI)
All Patients 9486 97.9% (97.5%, 98.3%)
CT No/Unknown 6928 98.2% (97.7%, 98.6%)
CT Yes 2558 97.3% (96.3%, 98.0%)
RS < 11 1734 99.2% (98.3%, 99.6%)
CT No/Unknown 1658 99.1% (98.2%, 99.6%)
CT Yes 76 100% (., .)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Size 3.1-4 cm

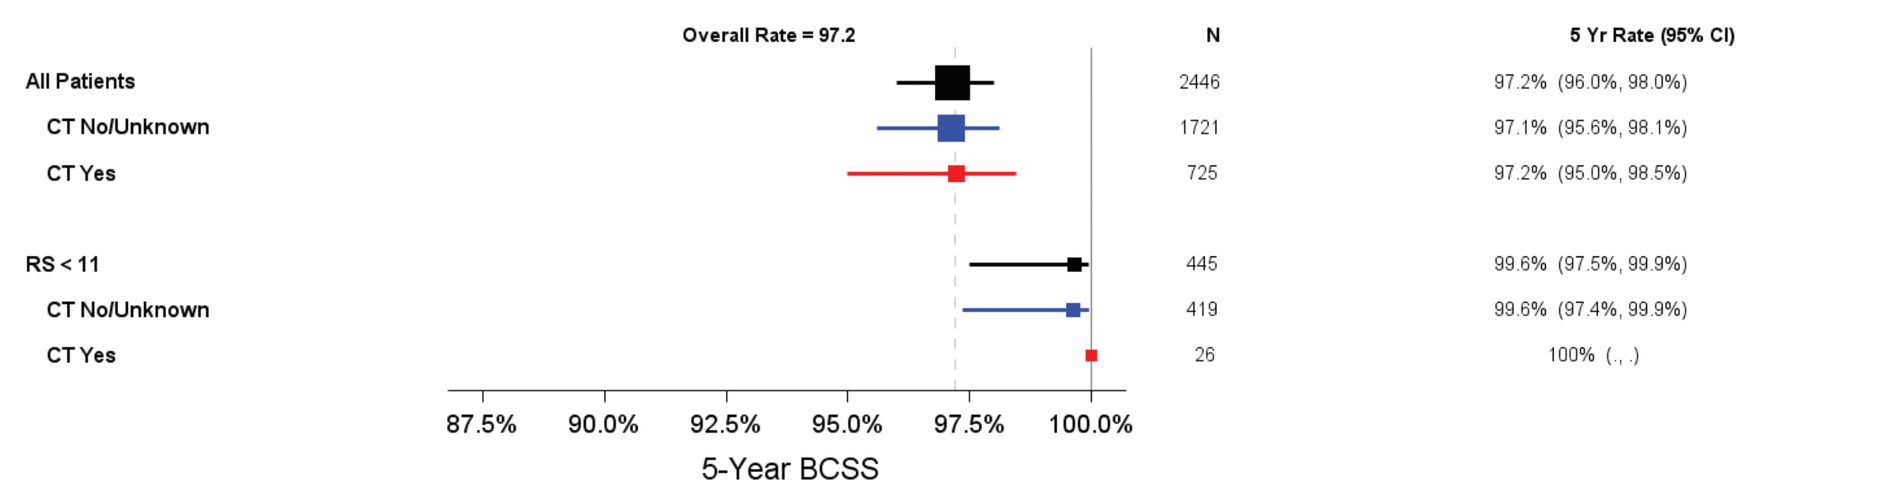
Overall Rate = 97.2
N
5 Yr Rate (95% CI)
All Patients 2446 97.2% (96.0%, 98.0%)
CT No/Unknown 1721 97.1% (95.6%, 98.1%)
CT Yes 725 97.2% (95.0%, 98.5%)
RS < 11 445 99.6% (97.5%, 99.9%)
CT No/Unknown 419 99.6% (97.4%, 99.9%)
CT Yes 26 100% (., .)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Size ≥4 cm

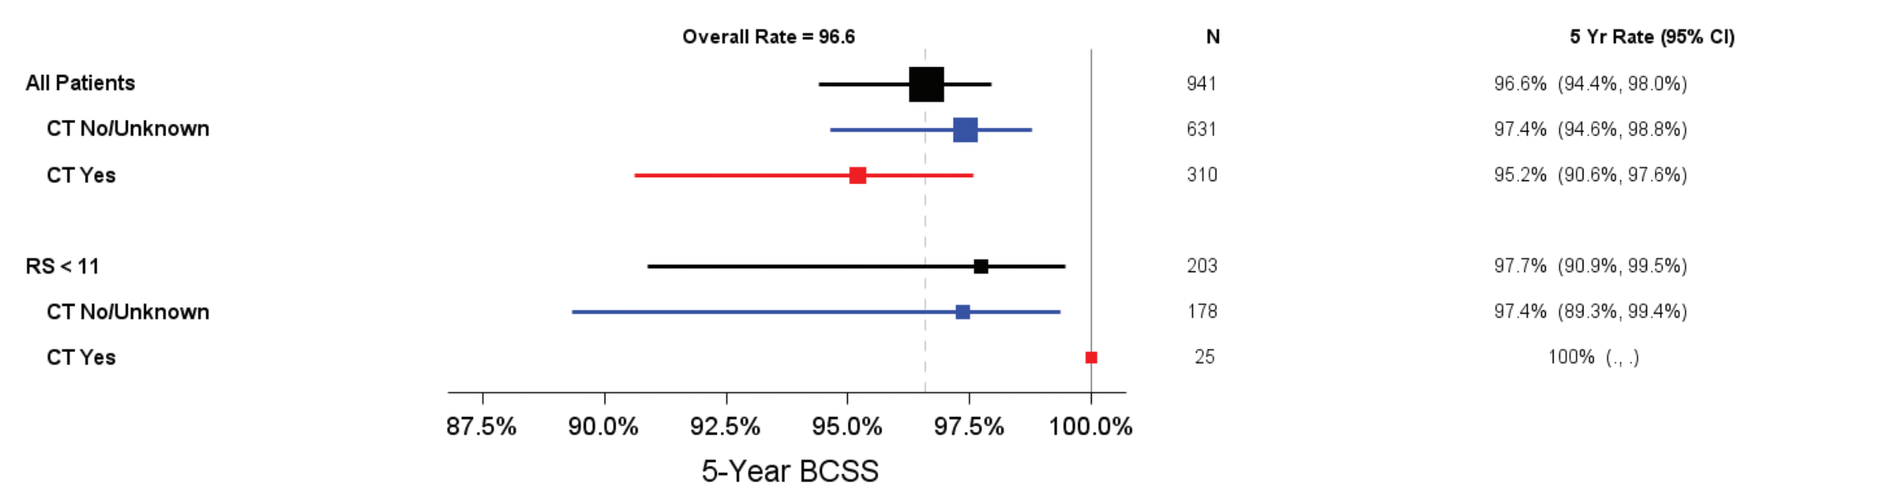
Overall Rate = 96.6
N
5 Yr Rate (95% CI)
All Patients 941 96.6% (94.4%, 98.0%)
CT No/Unknown 631 97.4% (94.6%, 98.8%)
CT Yes 310 95.2% (90.6%, 97.6%)
RS < 11 203 97.7% (90.9%, 99.5%)
CT No/Unknown 178 97.4% (89.3%, 99.4%)
CT Yes 25 100% (., .)
87.5% 90.0% 92.5% 95.0% 97.5% 100.0%
5-Year BCSS

Strengths and Limitations

STRENGTHS
- SEER rigor in cancer ascertainment and survival outcomes
- Largest cohort of patients with both traditional clinicopathologic factors assessed locally and 21-gene Oncotype DX Breast Recurrence Score assay results
- Population-based

LIMITATIONS
- Patients treated in clinical practice, no randomization to treatment
- SEER at this time has no information on recurrences.
- No central ER, PR, HER2 IHC, or HER2 FISH assessments

Summary and Conclusions

- The 9,000 patients with RS <11 treated with hormonal therapy alone had 99.6% 5-year BCSS
 - Excellent 5-year outcomes also observed with RS <11 in young patients, and in patients with large or high-grade tumors
- SEER real-world evidence of outcomes of patients treated based on RS results confirm new AJCC 8th edition staging criteria that many with poor prognostic factors may be down staged to Stage IA using the 21-gene assay and treated with hormonal therapy alone.

References

1. Hortobagyi GN, et al. Breast. In: Amin MB, et al., eds. *AJCC Cancer Staging Manual*. 8th ed. New York, NY: Springer Publishing; 2017:589-628.
2. Sparano JA, et al. *N Engl J Med*. 2015;373(21):2005-14.
3. SEER. https://seer.cancer.gov/
4. Petkov VI, et al. *npj Breast Cancer.* 2016;2:16017.
5. Miller DP, et al. *J Clin Oncol.* 2017;35(suppl):abstract 537.
6. Noone AM, et al. *Med Care.* 2016;54(9):e55-64.
7. Howlader N, et al. *J Natl Cancer Inst.* 2010;102(20):1584-98.

The authors thank John Bennett for biostatistics and graphics support, and Stan Mims for help with poster layout.
Oncotype DX, Recurrence Score, Oncotype DX Breast Recurrence Score, and Genomic Health are registered trademarks of Genomic Health, Inc.
This presentation is the intellectual property of the authors. Contact Dr. Baehner at <rbaehner@genomichealth.com> for permission to reprint and/or distribute.
Presented at: 40th Annual SABCS; December 5-9, 2017; San Antonio, TX. Poster P1-06-03.